In Vitro Evaluation of Naltrexone HCl 1% Topical Cream in Xematop for Psoriasis

Guiyun Song[1*], Kendice Ip[1], Daniel Banov[1], August S. Bassani[1], Benigno C. Valdez[2]

1. PCCA, Houston (TX) USA

2. Department of Stem Cell Transplantation & Cellular Therapy, The University of Texas MD Anderson Cancer Center, Houston (TX) USA

Introduction

Psoriasis is a multifactorial skin disease involving abnormal cell proliferation and inflammation. The plethora of treatment options available to patients may not be tolerable, efficacious, or affordable, and the development of therapeutic measures for psoriasis remains a challenge. In the search for more efficacious and convenient treatment for psoriasis, we developed a topical cream containing low-dose naltrexone (LDN) and a new compounding base named xematop. This base was specifically developed for topical formulations for skin conditions as it contains natural boswellic acid, avenanthramides from oats, phosphatidylglycerol and elegant film formers, which facilitate delivery of common active pharmaceutical ingredients, nourish and replenish the lamellar bilayers of the skin, restore health into the skin's barrier that may prevent water loss, and reduce skin redness and irritation [1,2].

In the present study, we report the biochemical evaluation of a new compounded formulation of xematop containing 1% naltrexone HCl, using a 3-dimensional (3D) reconstructed psoriasis tissue model. A stability study was also performed to evaluate the physical properties of this formulation and the chemical stability of naltrexone 1% strength. Our results provide a basis for clinical evaluation of this new formulation in managing the symptoms of psoriasis [1].

Materials and Methods

A three-dimensional psoriasis tissue model (SOR-300-FT) from MatTek Corp. (Ashland, MA) was exposed to the formulation for 2 or 5 days and analyzed for the level of markers of cellular proliferation, and inflammatory cytokine IL-6 [1].

- **Immunohistochemical (IHC) staining**

Tissue samples were harvested on day 2 for Ki67 expression analysis by IHC staining and analysis was performed using the two-step staining method. The slides were stained with 3,3-diaminobenzidine at room temperature for 2 min to detect the antigen and dehydrated with an ascending series of alcohols prior to mounting. Proliferating cells were stained brown and digital images were taken at 10X magnification [1].

- **Western blot analysis**

Western blot analysis was done by separating protein extracts (20 µg) on poly-acrylamide-SDS gels and blotting onto nitrocellulose membranes (Bio-Rad Laboratories, Hercules, CA). The primary antibodies used were raised against human PCNA, CYCLIN D1, β-CATENIN, c-MYC, c-JUN, MET, PI3K p85, AKT, mTOR, and β-ACTIN. Immunoblot analysis by chemiluminescence was done using the Immobilon Western Chemiluminescent Substrate [1].

- **ELISA assay for IL-6**

Psoriasis tissue growth media were collected on day 5 for Enzyme-Linked Immunosorbent Assay (ELISA) detection of IL-6 and Ki67. Samples and biotinylated primary antibody were mixed in the provided 96-well plate, which was then covered and incubated at room temperature for 1 hr, added streptavidin-HRP solution and chromogen 3,3',5,5'-tetramethylbenzidine substrate solution; optical density was measured at 450 nm [1].

- **Physical and chemical stability**

For physicochemical stability analysis, 14 plastic jars (25 g) were prepared. Half of the preparations was stored at refrigerated temperature (5°C) and the other half was stored at controlled room temperature (25°C) with 60% relative humidity. At 0, 7, 21, 30, 43, 61, 90 and 180 days, one unopened jar was withdrawn and analyzed for changes in color/appearance, viscosity, and pH. The stability-indicating UPLC method used to analyze naltrexone was developed to separate naltrexone from its degradation products and it was validated for specificity, linearity, system suitability, accuracy, precision, robustness and solution stability. Chemical stability was defined as the retention of 90% to 110% of the initial concentration [1].

77TH ANNUAL MEETING
SID SOCIETY FOR INVESTIGATIVE DERMATOLOGY 1937
CHICAGO 2019 MAY 8-11, 2019 HILTON CHICAGO

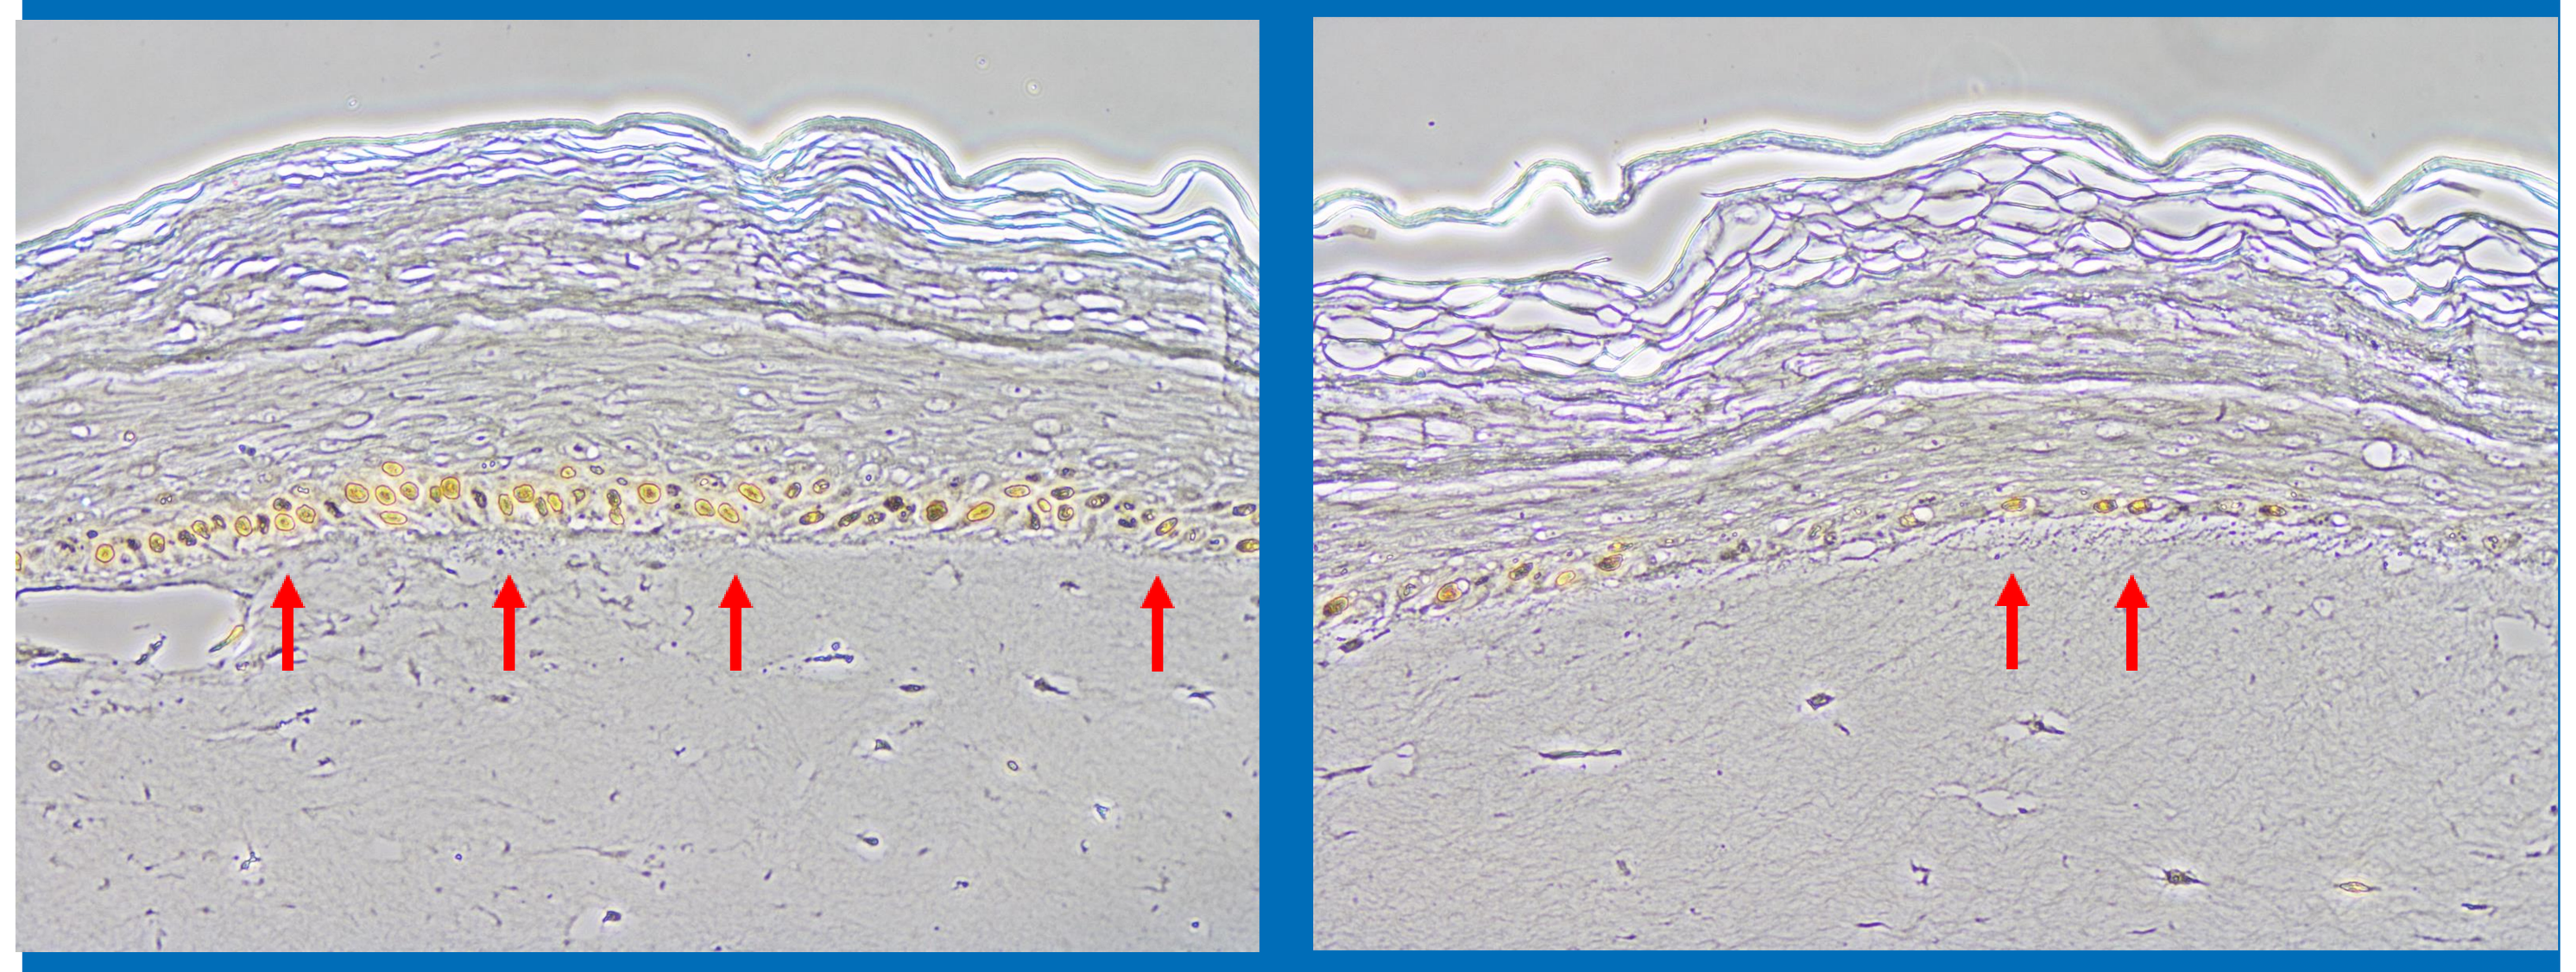

Figure 1. ***Psoriasis tissues treated with naltrexone 1% in xematop for 2 days (right) and control tissues (left). The red arrows indicate the stained proliferation marker Ki67.***

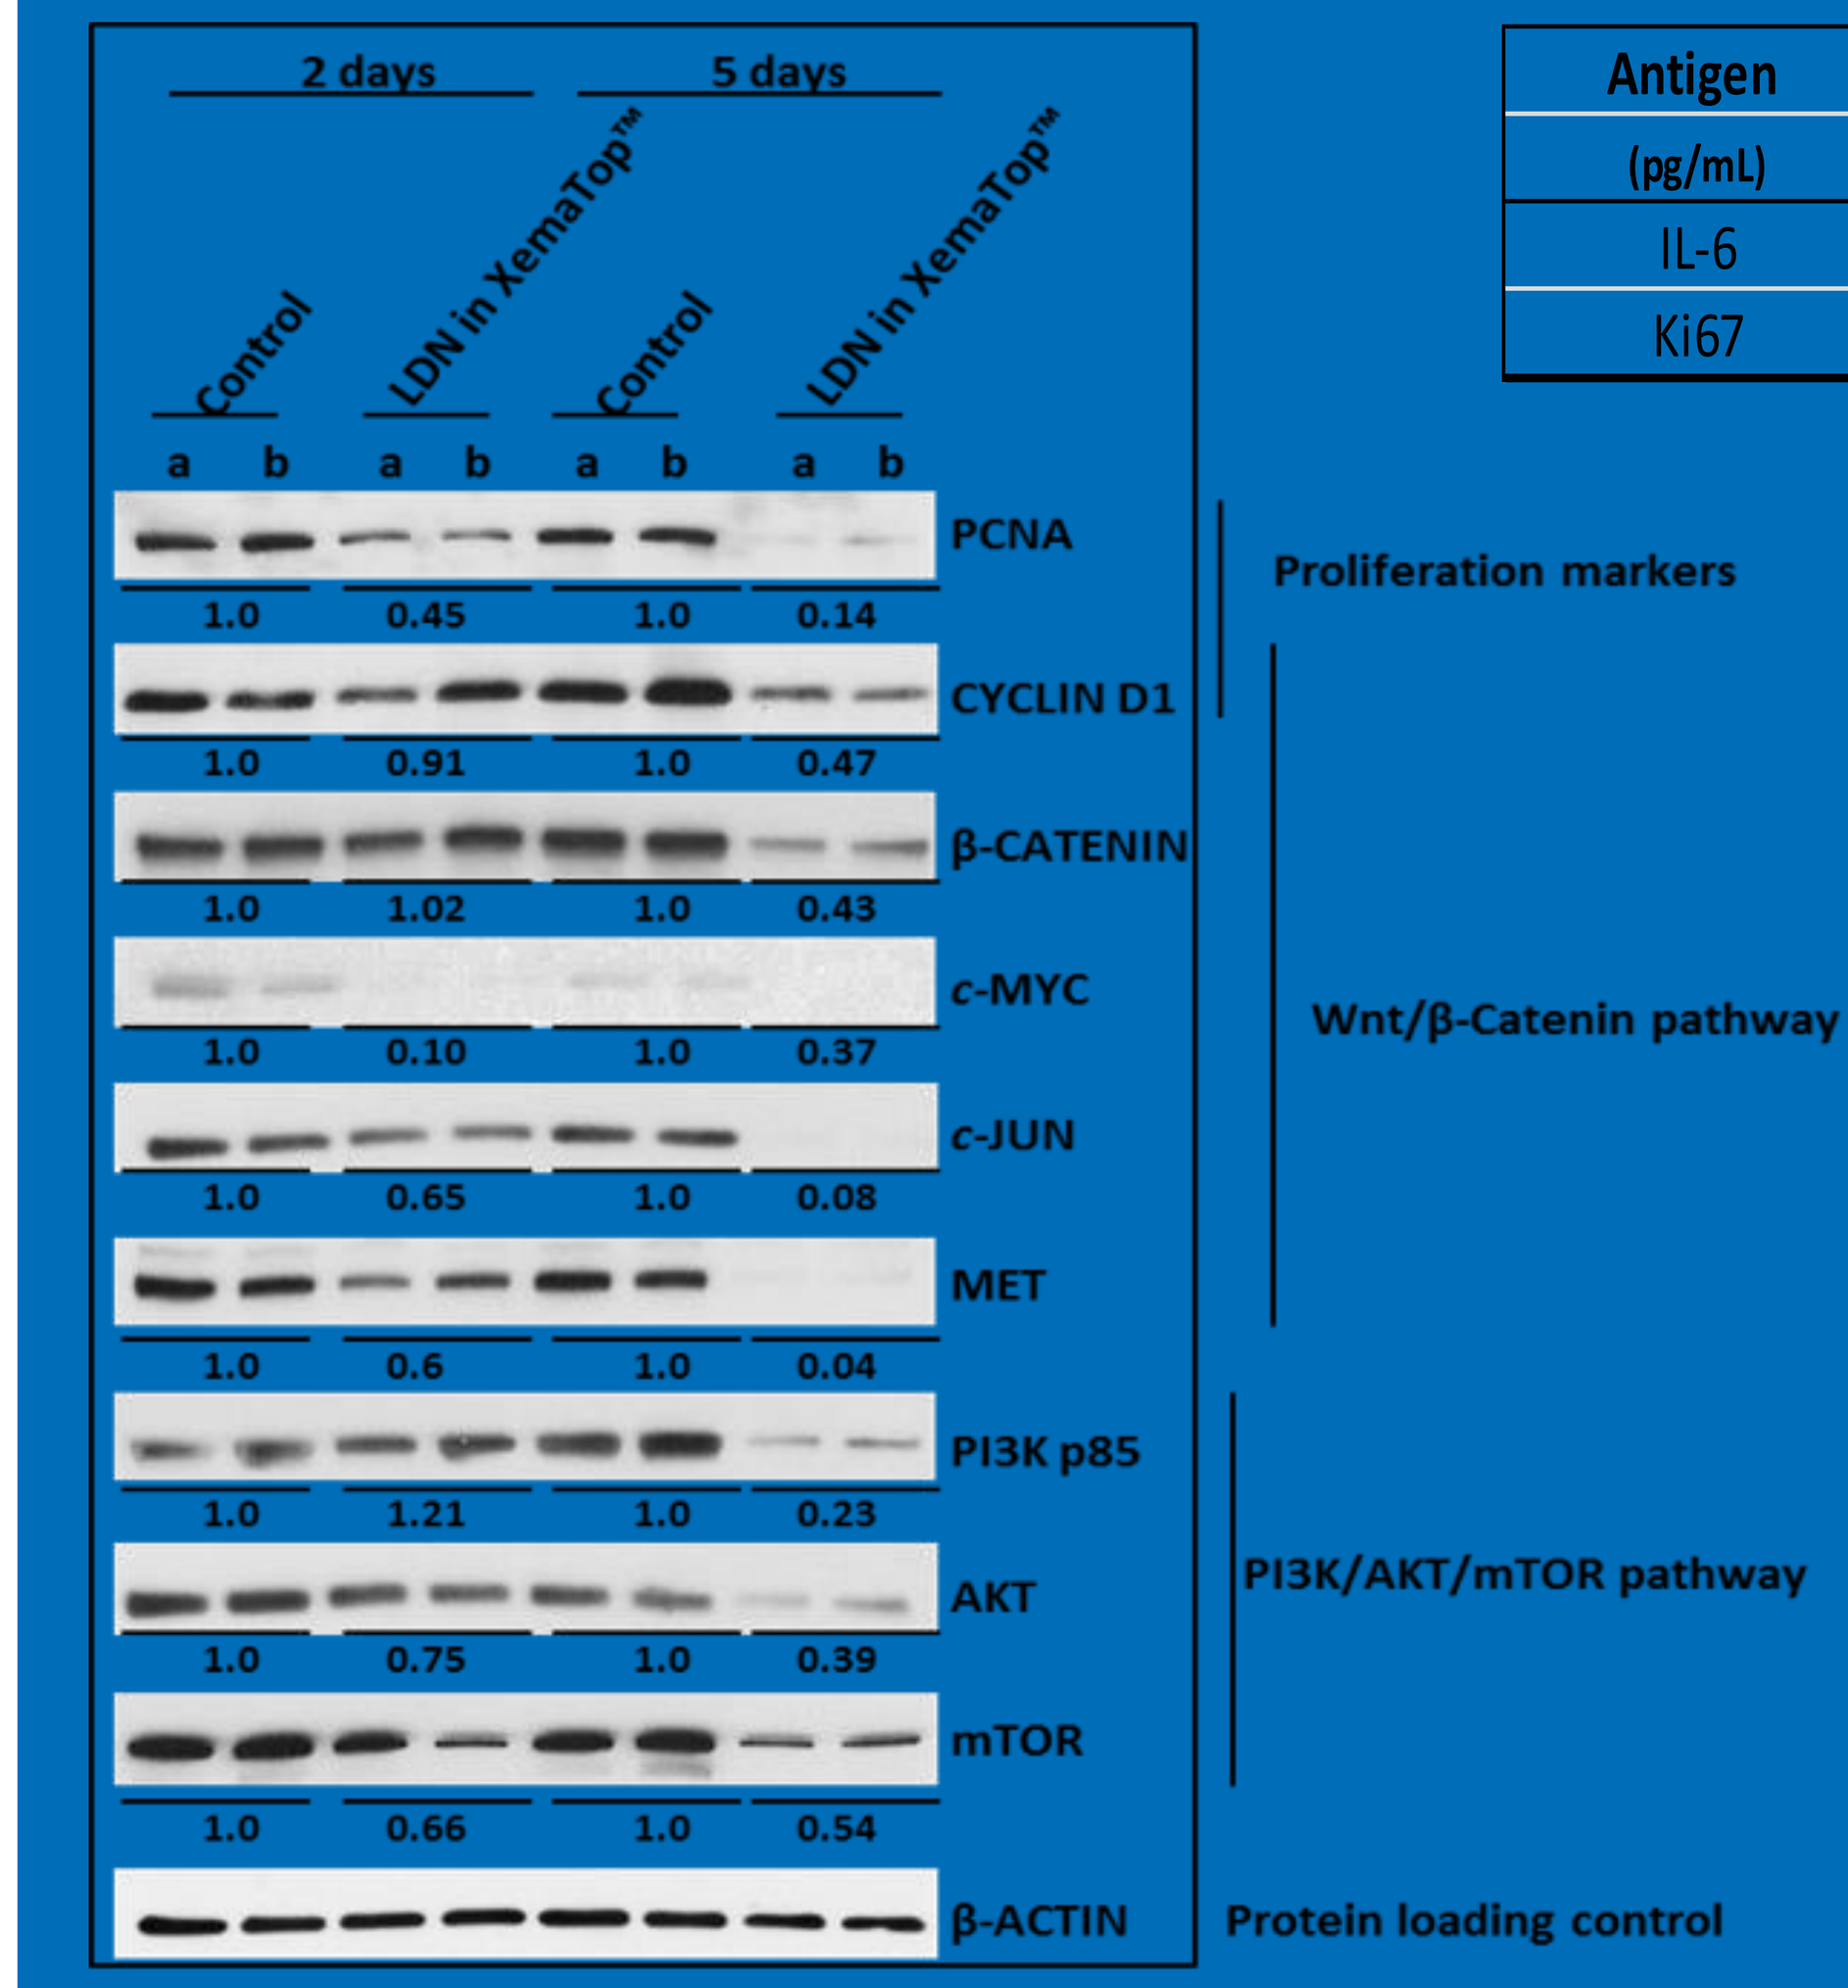
2 days
5 days
Control
LDN in XemaTop™
Control
LDN in XemaTop™
a b a b a b a b
PCNA 1.0 0.45 1.0 0.14
CYCLIN D1 1.0 0.91 1.0 0.47
β-CATENIN 1.0 1.02 1.0 0.43
c-MYC 1.0 0.10 1.0 0.37
c-JUN 1.0 0.65 1.0 0.08
MET 1.0 0.6 1.0 0.04
PI3K p85 1.0 1.21 1.0 0.23
AKT 1.0 0.75 1.0 0.39
mTOR 1.0 0.66 1.0 0.54
β-ACTIN
Proliferation markers
Wnt/β-Catenin pathway
PI3K/AKT/mTOR pathway
Protein loading control

Antigen (pg/mL)	Control	LDN in XemaTop™	*P* value
IL-6	166.3 ± 12.2	48.6 ± 9.8	0.000006
Ki67	2.1 ± 0.2	0.8 ± 0.2	0.02

Figure 2. ***Western blot analysis: total cell extracts from psoriasis tissue model treated with the compounded drug (left). ELISA analysis: Culture media from tissues exposed to compounded drug (right).***

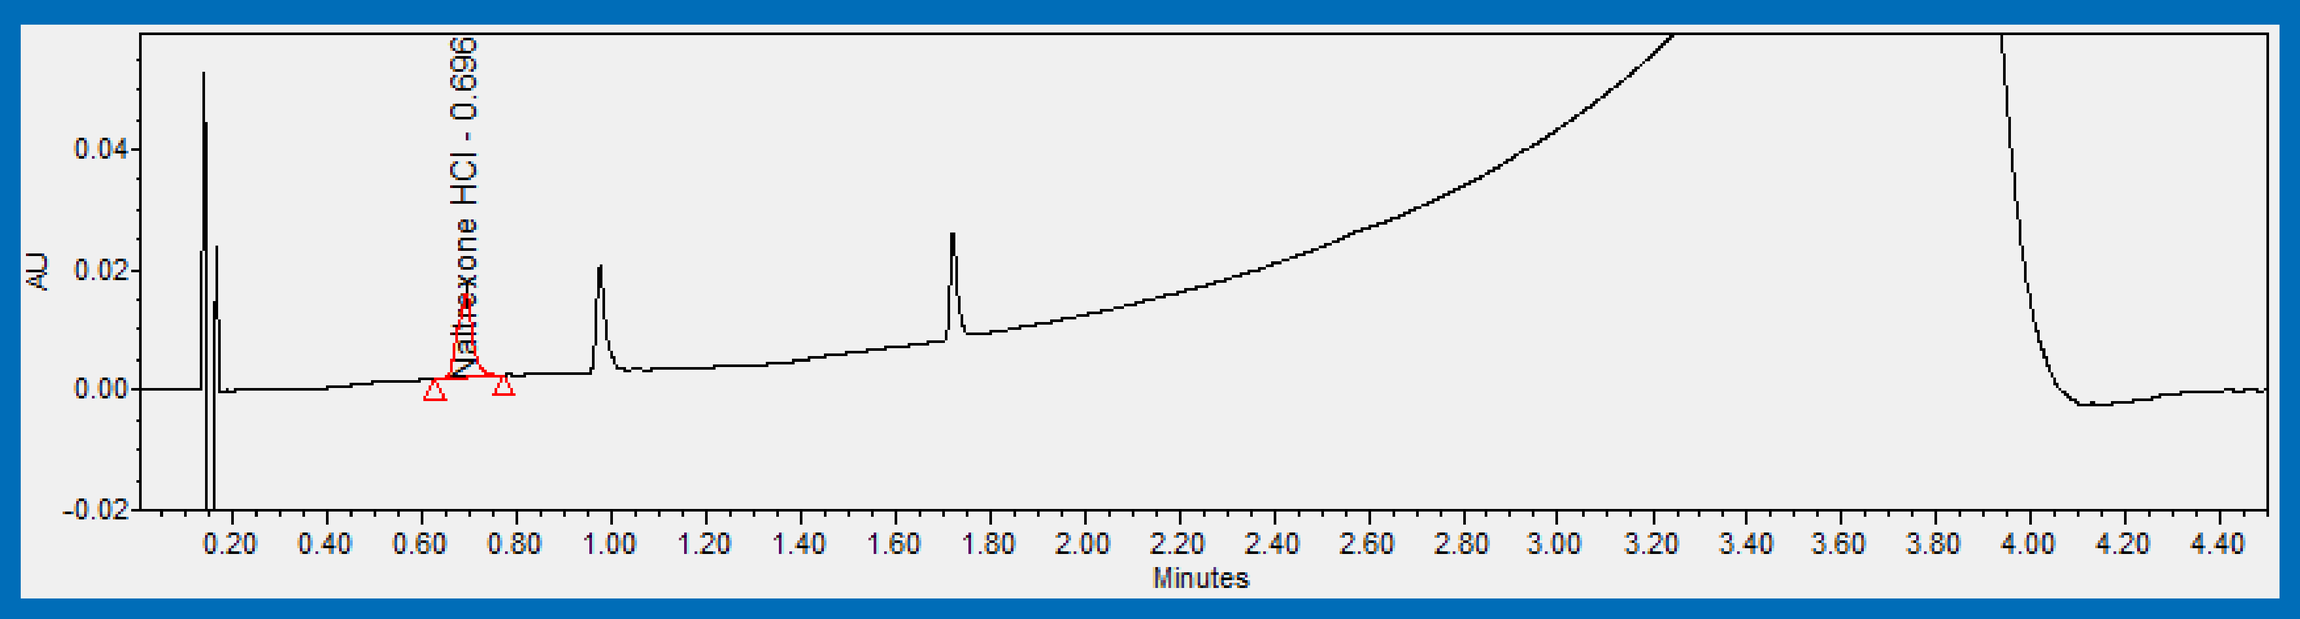
Naltrexone HCl - 0.696
AU
Minutes

Figure 3. ***Sample UPLC chromatograms: naltrexone HCl 1% topical cream in xematop.***

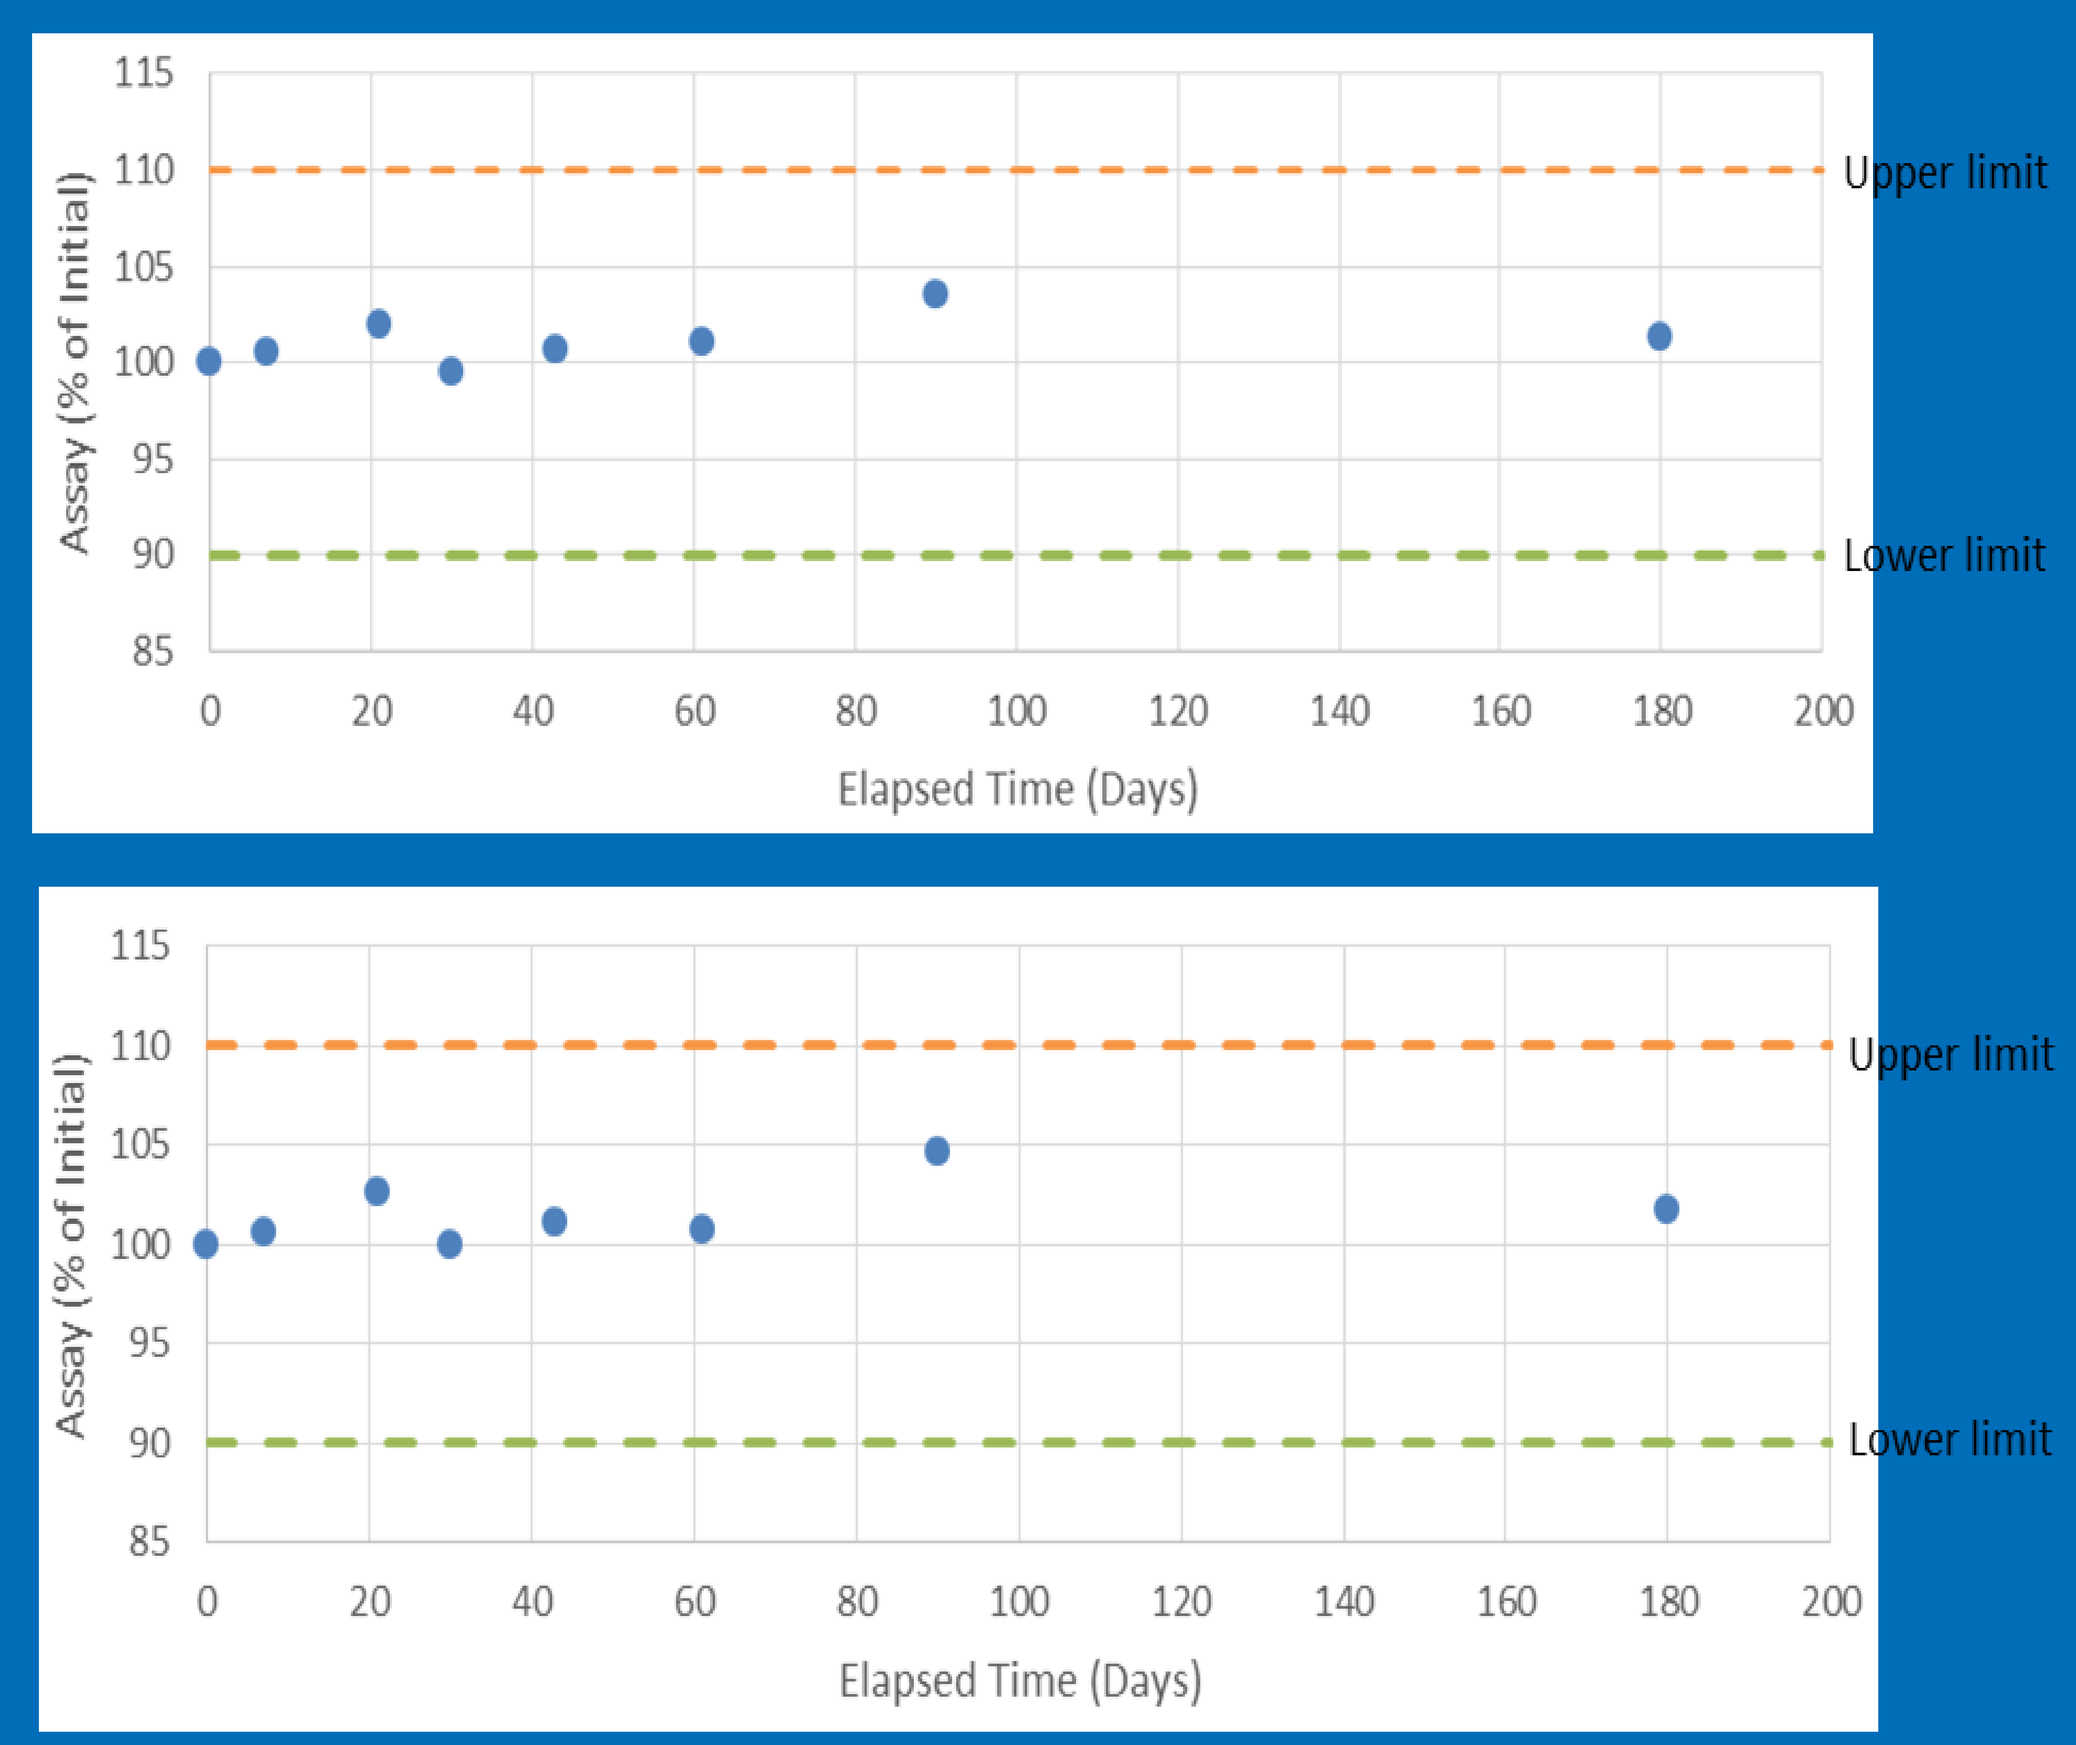
Upper limit
Lower limit
Assay (% of Initial)
Elapsed Time (Days)
Upper limit
Lower limit
Assay (% of Initial)
Elapsed Time (Days)

Figure 4. ***Stability of Naltrexone HCl 1% in xematop. The compounded formulation was stored at refrigerated (5°C, top) or room (25°C, bottom) temperature and analyzed for the level of naltrexone by UPLC.***

Results and Discussion

- **1% naltrexone HCl in xematop inhibits cellular proliferation and production of IL-6 in psoriasis tissue model.**

Using immunohistochemical staining, the level of Ki67 protein significantly decreased in the drug-treated tissues. Figure 1 shows a significant decrease in the brown staining of cells (indicated by red arrows) located between the dermis and epidermis layers, showing decreased expression of Ki67 and suggesting decreased proliferation of keratinocytes. The exact mechanism on how LDN inhibits proliferation of keratinocytes remains to be determined. Western blot analysis showed 86% and 53% down-regulation of other proliferation markers PCNA and CYCLIN D1, respectively, after 5-day exposure. The pro-survival Wnt/β-catenin pathway was compromised as indicated by 57% decrease in the level of β-CATENIN and down-regulation of its downstream targets including CYCLIN D1 (decreased by 53%), c-MYC (63%), c-JUN (92%) and MET (96%) proteins. Likewise, the PI3K/AKT/mTOR pathway was significantly inhibited by 1% naltrexone HCl in xematop.

The production of IL-6 was also inhibited by 70% in drug-treated tissues exposed to the formula after 5 days (Figure 2). This decrease is consistent with the efficacy of some topical therapies on psoriasis in decreasing the levels of cytokines. The results of this study suggest that the new compounded formulation containing 1% naltrexone may be efficacious in alleviating the symptoms of psoriasis by inhibiting proliferation of keratinocytes and decreasing the levels of inflammatory cytokines [1].

- **1% naltrexone HCl in xematop is physically and chemically stable.**

The appearance and color of the compounded formulation remained smooth with a faint beige color throughout the study period of 180 days under both refrigerated and room temperature storage conditions. No malodor was observed from the samples. The measured pH values did not increase or drop significantly, indicating pH stability of the formulation [1].

The level of naltrexone was quantified using a validated stability-indicating UPLC assay method as all criteria were met. According to the sample UPLC chromatogram shown in Figure 3, using a standard naltrexone solution, the main peak appeared at around 0.696 min. However, additional peaks at 0.979 min and 1.723 min were observed in the placebo and the compounded formulation, which were considered components in excipients. There was no interference to the main naltrexone peak from degradant or component in the excipient. Naltrexone degraded for about 27% when exposed to heat at 60°C for three days. Having the UPLC method validated for its specificity, it was then used to determine the chemical stability of naltrexone in xematop. The initial concentration was set as the baseline for percent remaining in the duration of the study. The percent remaining of the preparation was within 99.6% to 103.6% from day 7 to 180 when stored at refrigerated temperature, and was within 100.0% to 104.7% from day 7 to 180 when stored at controlled room temperature (Figure 4) [1].

The results of this study show that the newly compounded formulation in plastic jars at refrigerated and room temperatures demonstrated physical and chemical stability for up to 180 days. These physicochemical stability data could be used to assign the BUD for the formula [1].

Conclusions

Xematop is non-toxic to human skin cells. Using it as a base for 1% naltrexone may provide a topical treatment option for psoriasis patients. The observed decrease in the level of psoriasis biomarkers suggests that the compounded formula may attenuate the inflammatory response and cellular proliferation associated with psoriasis in vivo. Moreover, xematop with 1% naltrexone demonstrated physicochemical stability for up to 180 days at both refrigerated and room temperatures, which can be used to assign BUD and intended storage conditions [1].

*Correspondence to: gsong@pccarx.com

[1] Ip, K., Song, G., Banov, D., Bassani, A.S. and Valdez, B.C. (submitted) "*In vitro* evaluation of Naltrexone HCl 1% Topical Cream in XemaTop™ for psoriasis", *Archives of Dermatological Research*

[2] Professional Compounding Centers of America. XemaTop™. [PCCA Website.] Available at: http://www.pccarx.com/compounding-for-winter-conditions

Scan me

For a copy of this poster, please scan the QR code.